A Phase Ib Trial of the Anti - PD - L1 Human Monoclonal Antibody, CS1001, in Patients with Advanced Solid Tumors or Lymphomas: Preliminary Efficacy and Safety Results from 4 Cohorts

BARCELONA 2019 ESMO congress

1268P

Shen, Lin[1]; Li, Jin[2]; Xing, Baocai[3]; Xu, Nong[4]; Miao, Zhanhui[5]; Zhang, Qingyuan[6]; Zhao, Yanqiu[7]; Ding, Jiyuan[8]; Wang, Jingru[8]; Dai, Hangjun[8]; Wang, Yin[8]; Shi, Qingmei[8]; Yang, Jason[8]

1. Gastrointestinal Oncology, Peking University Cancer Hospital & Institute, Beijing, China 2. Oncology, Shanghai East Hospital, Tongji University School of Medicine, Shanghai, China 3. Hepatobiliary and Pancreatic Surgery, Beijing Cancer Hospital, Beijing, China 4. Medical Oncology, The First Affiliated Hospital of Zhejiang University, Hangzhou, China 5. Cancer Department of First Affiliated Hospital of Xinxiang Medical College, Xinxiang, China 6. Medical Oncology, Harbin Medical University Cancer Hospital, Harbin, China 7. Medical Oncology, Affiliated Cancer Hospital of Zhengzhou University, Henan Cancer Hospital, Zhengzhou, China 8. CStone Pharmaceuticals (Suzhou) Co., Ltd., Suzhou, China

基石药业 CSTONE PHARMACEUTICALS

BACKGROUND

- CS1001 is the first full-length, fully human programmed death ligand-1 (PD-L1) targeted immunoglobin G4 (IgG4, s228p) monoclonal antibody (mAb) developed by the OMT transgenic rat platform. The platform mirrors natural IgG4 human antibody, which may potentially reduce the risk of immunogenicity and toxicity in patients (pts)
- In Phase Ia part of the first-in-human study of CS1001 (NCT03312842), 1200 mg fixed dose every 3 weeks (Q3W) was determined as the recommended Phase 2 dose (RP2D)[1]
- Phase Ib is the dose-expansion phase to further explore the efficacy and safety of CS1001 in multiple cohorts of selected tumor types. Herein, with the cut-off date of 01 Jul 2019, we present the safety data from Phase Ib and efficacy data from 4 cohorts as following:
 - CS1001 monotherapy of cholangiocarcinoma or gallbladder carcinoma (CC/GBC);
 - CS1001 monotherapy of high-microsatellite instability or mismatch repair deficient phenotype (MSI-H/dMMR) (≥2L);
 - CS1001 and chemotherapy combination as first line (1L) treatment in gastric or gastro-esophageal junction carcinoma (GC/GEJ);
 - CS1001 and chemotherapy combination as 1L treatment in esophageal squamous cell carcinoma (ESCC)

METHODS

Key Eligibility

Eastern Cooperative Oncology Group (ECOG) performance status of 0-1

CC/GBC: pts with histologically/cytologically diagnosed unresectable CC/GBC who failed standard treatment or are unable to tolerate or refuse the standard treatment

MSI-H/dMMR (≥2L): pts with inoperable or metastatic MSI-H/dMMR solid tumors who failed the previous-line treatment before enrollment and do not have a satisfactory alternative treatment

GC/GEJ (1L): pts with unresectable locally advanced or metastatic GC/GEJ which was histologically confirmed as adenocarcinoma, and who did not receive systemic treatment for advanced or metastatic disease

ESCC (1L): pts with histologically confirmed unresectable, recurrent or distantly metastatic locally advanced ESCC who did not receive systemic treatment

Figure 1 Study Design and Objectives

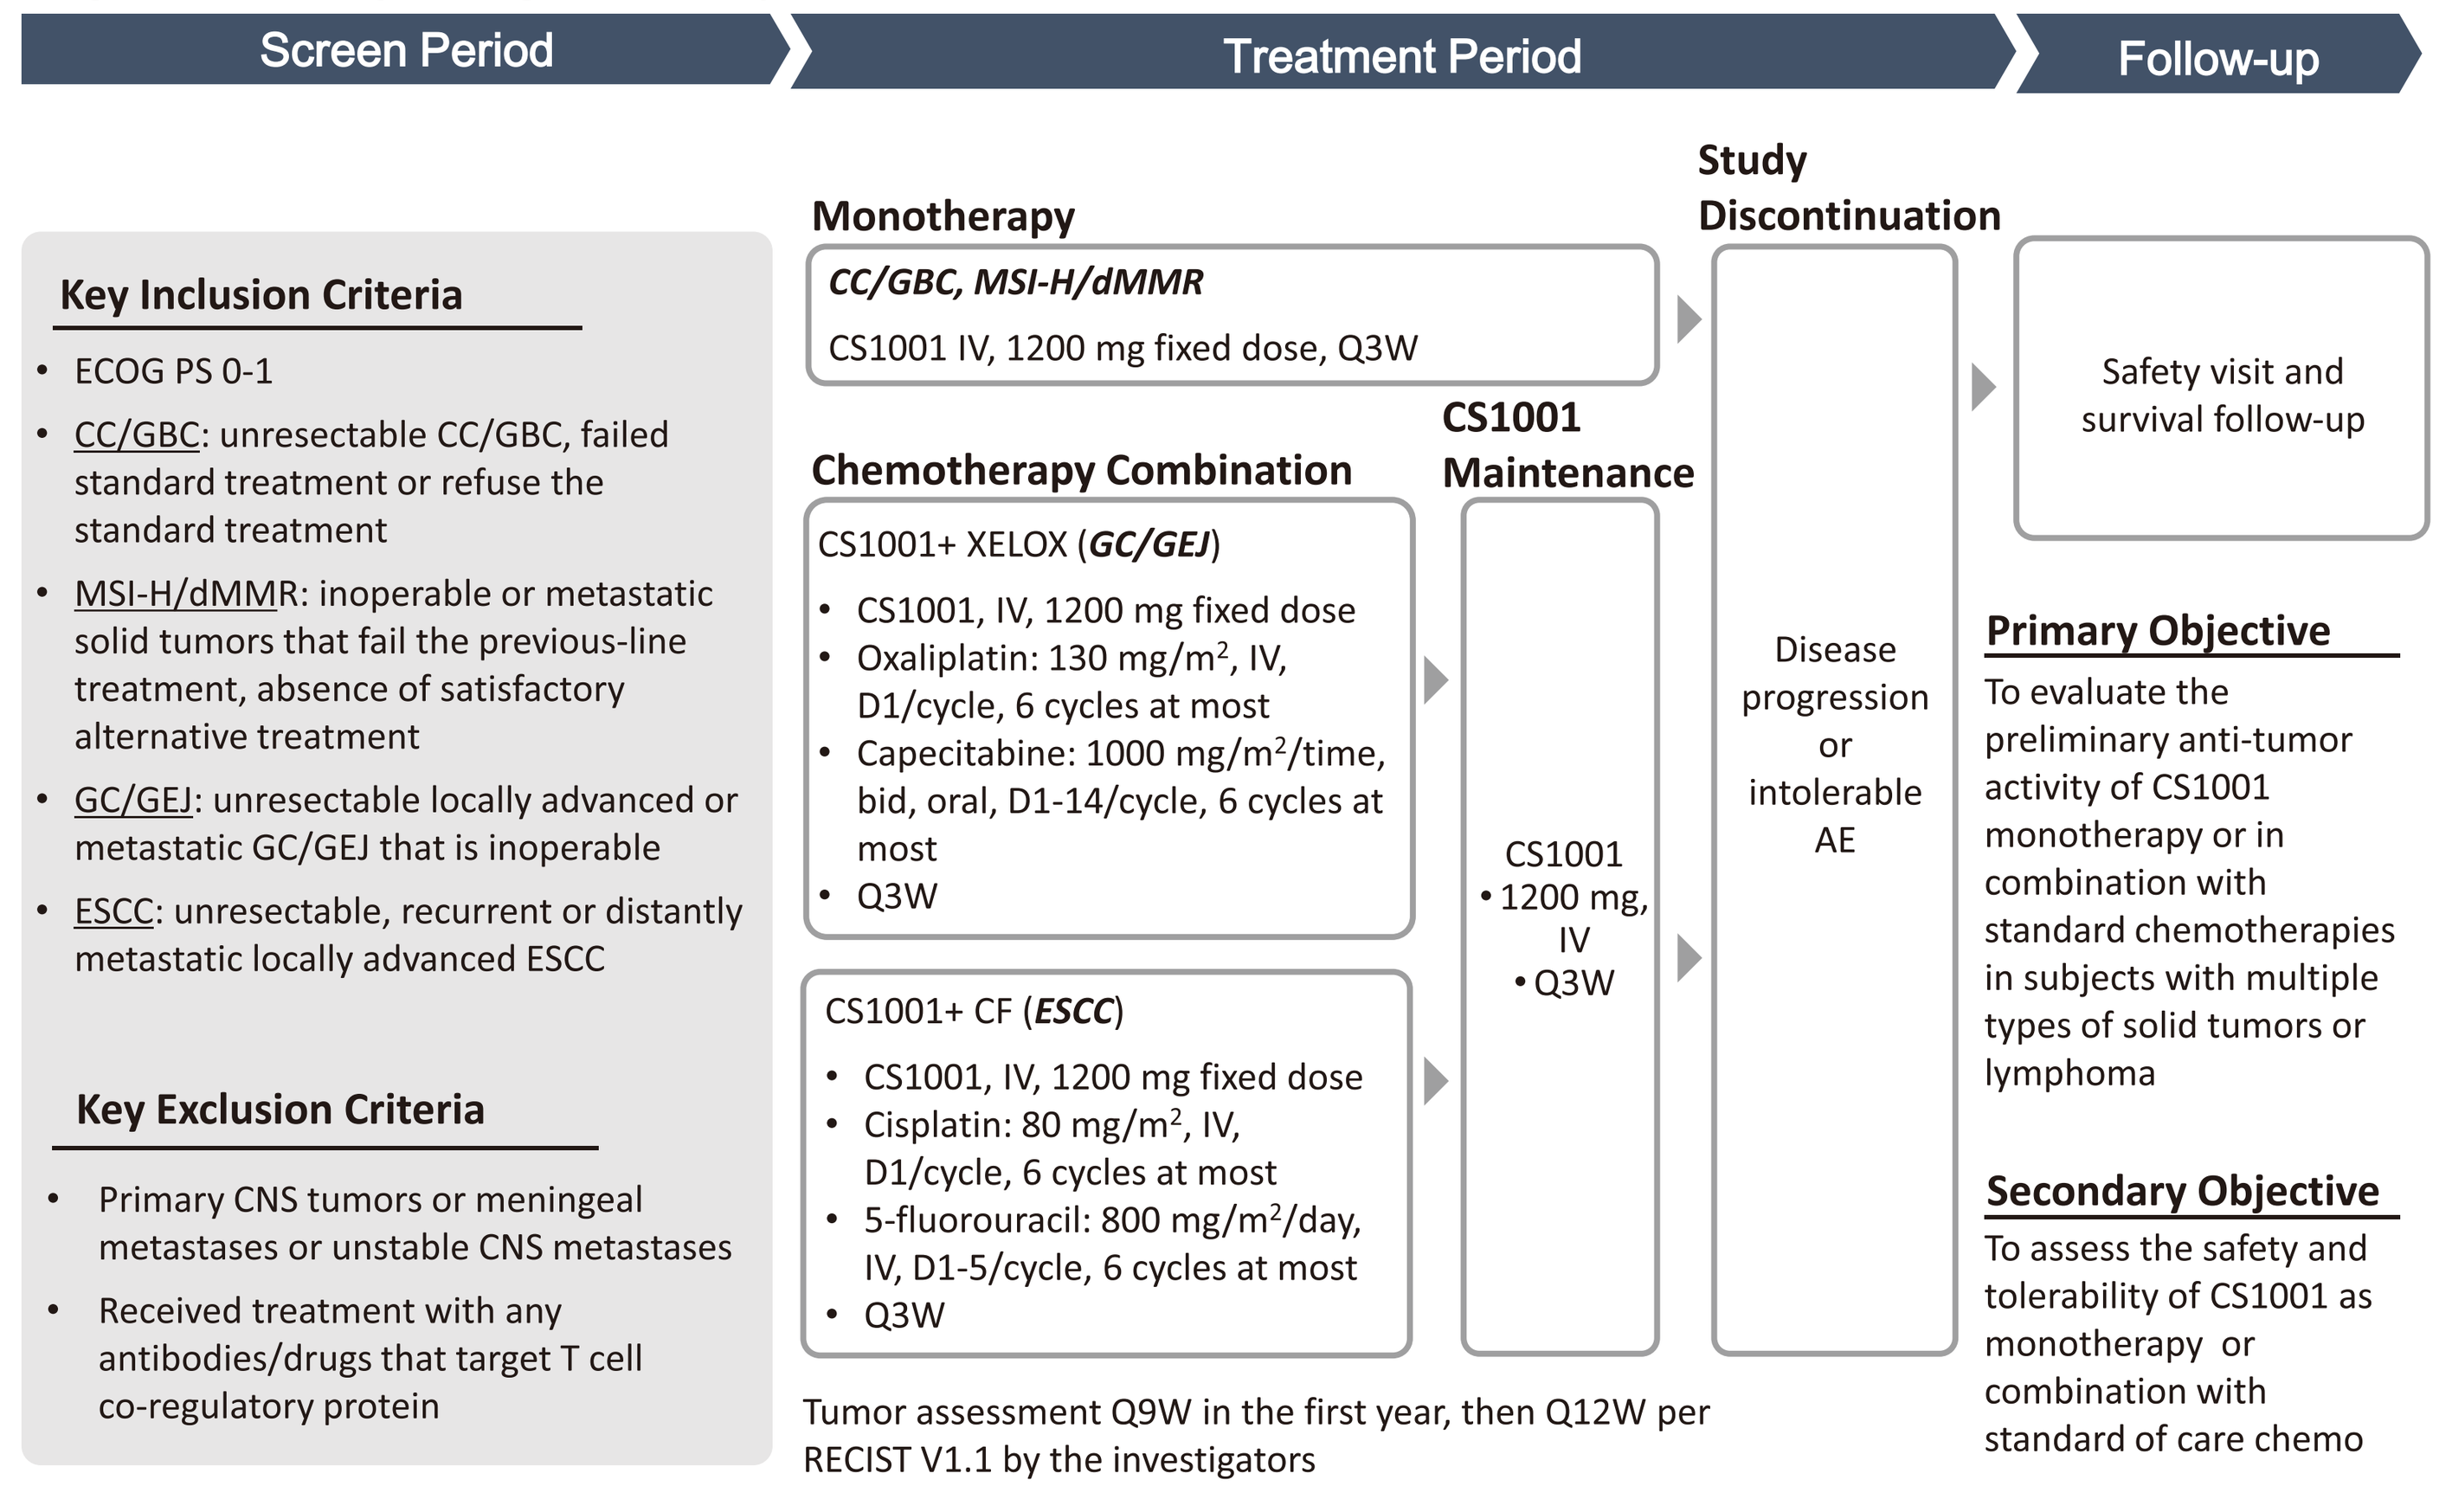
Screen Period
Treatment Period
Follow-up
Key Inclusion Criteria
• ECOG PS 0-1
• CC/GBC: unresectable CC/GBC, failed standard treatment or refuse the standard treatment
• MSI-H/dMMR: inoperable or metastatic solid tumors that fail the previous-line treatment, absence of satisfactory alternative treatment
• GC/GEJ: unresectable locally advanced or metastatic GC/GEJ that is inoperable
• ESCC: unresectable, recurrent or distantly metastatic locally advanced ESCC
Key Exclusion Criteria
• Primary CNS tumors or meningeal metastases or unstable CNS metastases
• Received treatment with any antibodies/drugs that target T cell co-regulatory protein
Monotherapy
CC/GBC, MSI-H/dMMR
CS1001 IV, 1200 mg fixed dose, Q3W
Chemotherapy Combination
CS1001+ XELOX (GC/GEJ)
• CS1001, IV, 1200 mg fixed dose
• Oxaliplatin: 130 mg/m2, IV, D1/cycle, 6 cycles at most
• Capecitabine: 1000 mg/m2/time, bid, oral, D1-14/cycle, 6 cycles at most
• Q3W
CS1001+ CF (ESCC)
• CS1001, IV, 1200 mg fixed dose
• Cisplatin: 80 mg/m2, IV, D1/cycle, 6 cycles at most
• 5-fluorouracil: 800 mg/m2/day, IV, D1-5/cycle, 6 cycles at most
• Q3W
CS1001 Maintenance
CS1001 • 1200 mg, IV • Q3W
Study Discontinuation
Disease progression or intolerable AE
Safety visit and survival follow-up
Primary Objective
To evaluate the preliminary anti-tumor activity of CS1001 monotherapy or in combination with standard chemotherapies in subjects with multiple types of solid tumors or lymphoma
Secondary Objective
To assess the safety and tolerability of CS1001 as monotherapy or combination with standard of care chemo
Tumor assessment Q9W in the first year, then Q12W per RECIST V1.1 by the investigators

AE: adverse event; bid: twice daily; CNS: central nervous system; D: day; IV: intravenous; ECOG: Eastern Cooperative Oncology Group; PS: performance status; Q3W: once every 3 weeks

Assessments

- Tumor assessment was assessed per RECIST V1.1 by investigators every 9 weeks (Q9W) in the first year, then every 12 weeks (Q12W)
- Adverse events (AEs) were graded according to the National Cancer Institute Common Terminology Criteria for Adverse Event (NCI-CTCAE) V4.03

RESULTS

Patients – 4 Cohorts

Table 1. Demographics and Baseline Characteristics (Safety Analysis Set)

Characteristics		Monotherapy		Chemotherapy Combination	
		CC/GBC N=29	MSI-H/dMMR (≥ 2L) N=21	GC/GEJ (1L) N=29	ESCC (1L) N=23
Age (years), Median (range)		55 (39, 72)	53 (25, 71)	60 (40, 73)	61 (45, 73)
Sex, n (%),	Male	9 (31.0)	12 (57.1)	23 (79.3)	18 (78.3)
	Female	20 (69.0)	9 (42.9)	6 (20.7)	5 (21.7)
ECOG PS, n (%)	0	14 (48.3)	0	12 (41.4)	5 (21.7)
	1	13 (44.8)	21 (100)	17 (58.6)	18 (78.3)
	Missing	2 (6.9)	0	0	0
Time since Initial Diagnosis (years)	Median (range)	0.731 (0.03, 1.84)	1.320 (0.36, 19.76)	0.077 (0.01, 6.60)	0.487 (0.01, 3.04)
Prior Anti-Cancer Therapy Regimen	Median (range)	1.0 (0, 6)	2.0 (1, 9)	0.0 (0, 2)*	0.0 (0, 2)**
Current Cancer Stage, n (%)	Stage III	0	0	1 (3.4)	3 (13)
	Stage IV	29 (100)	21 (100)	28 (96.6)	20 (87)

ECOG: Eastern Cooperative Oncology Group; PS: performance status
*A total of 10 patients received neoadjuvant and/or adjuvant therapy. 1 patient received both neo-adjuvant and adjuvant therapies around curative surgery.
**A total of 8 patients received adjuvant and/or neoadjuvant drug therapy, among which 1 patient received cisplatin + paclitaxel after radical surgery, and subsequently changed to cisplatin + capecitabine due to taxol allergy.

Efficacy – 4 Cohorts

Figure 2 Waterfall Plot of Maximum Target Lesion Shrinkage by RECIST V1.1 (Efficacy Analysis Set)

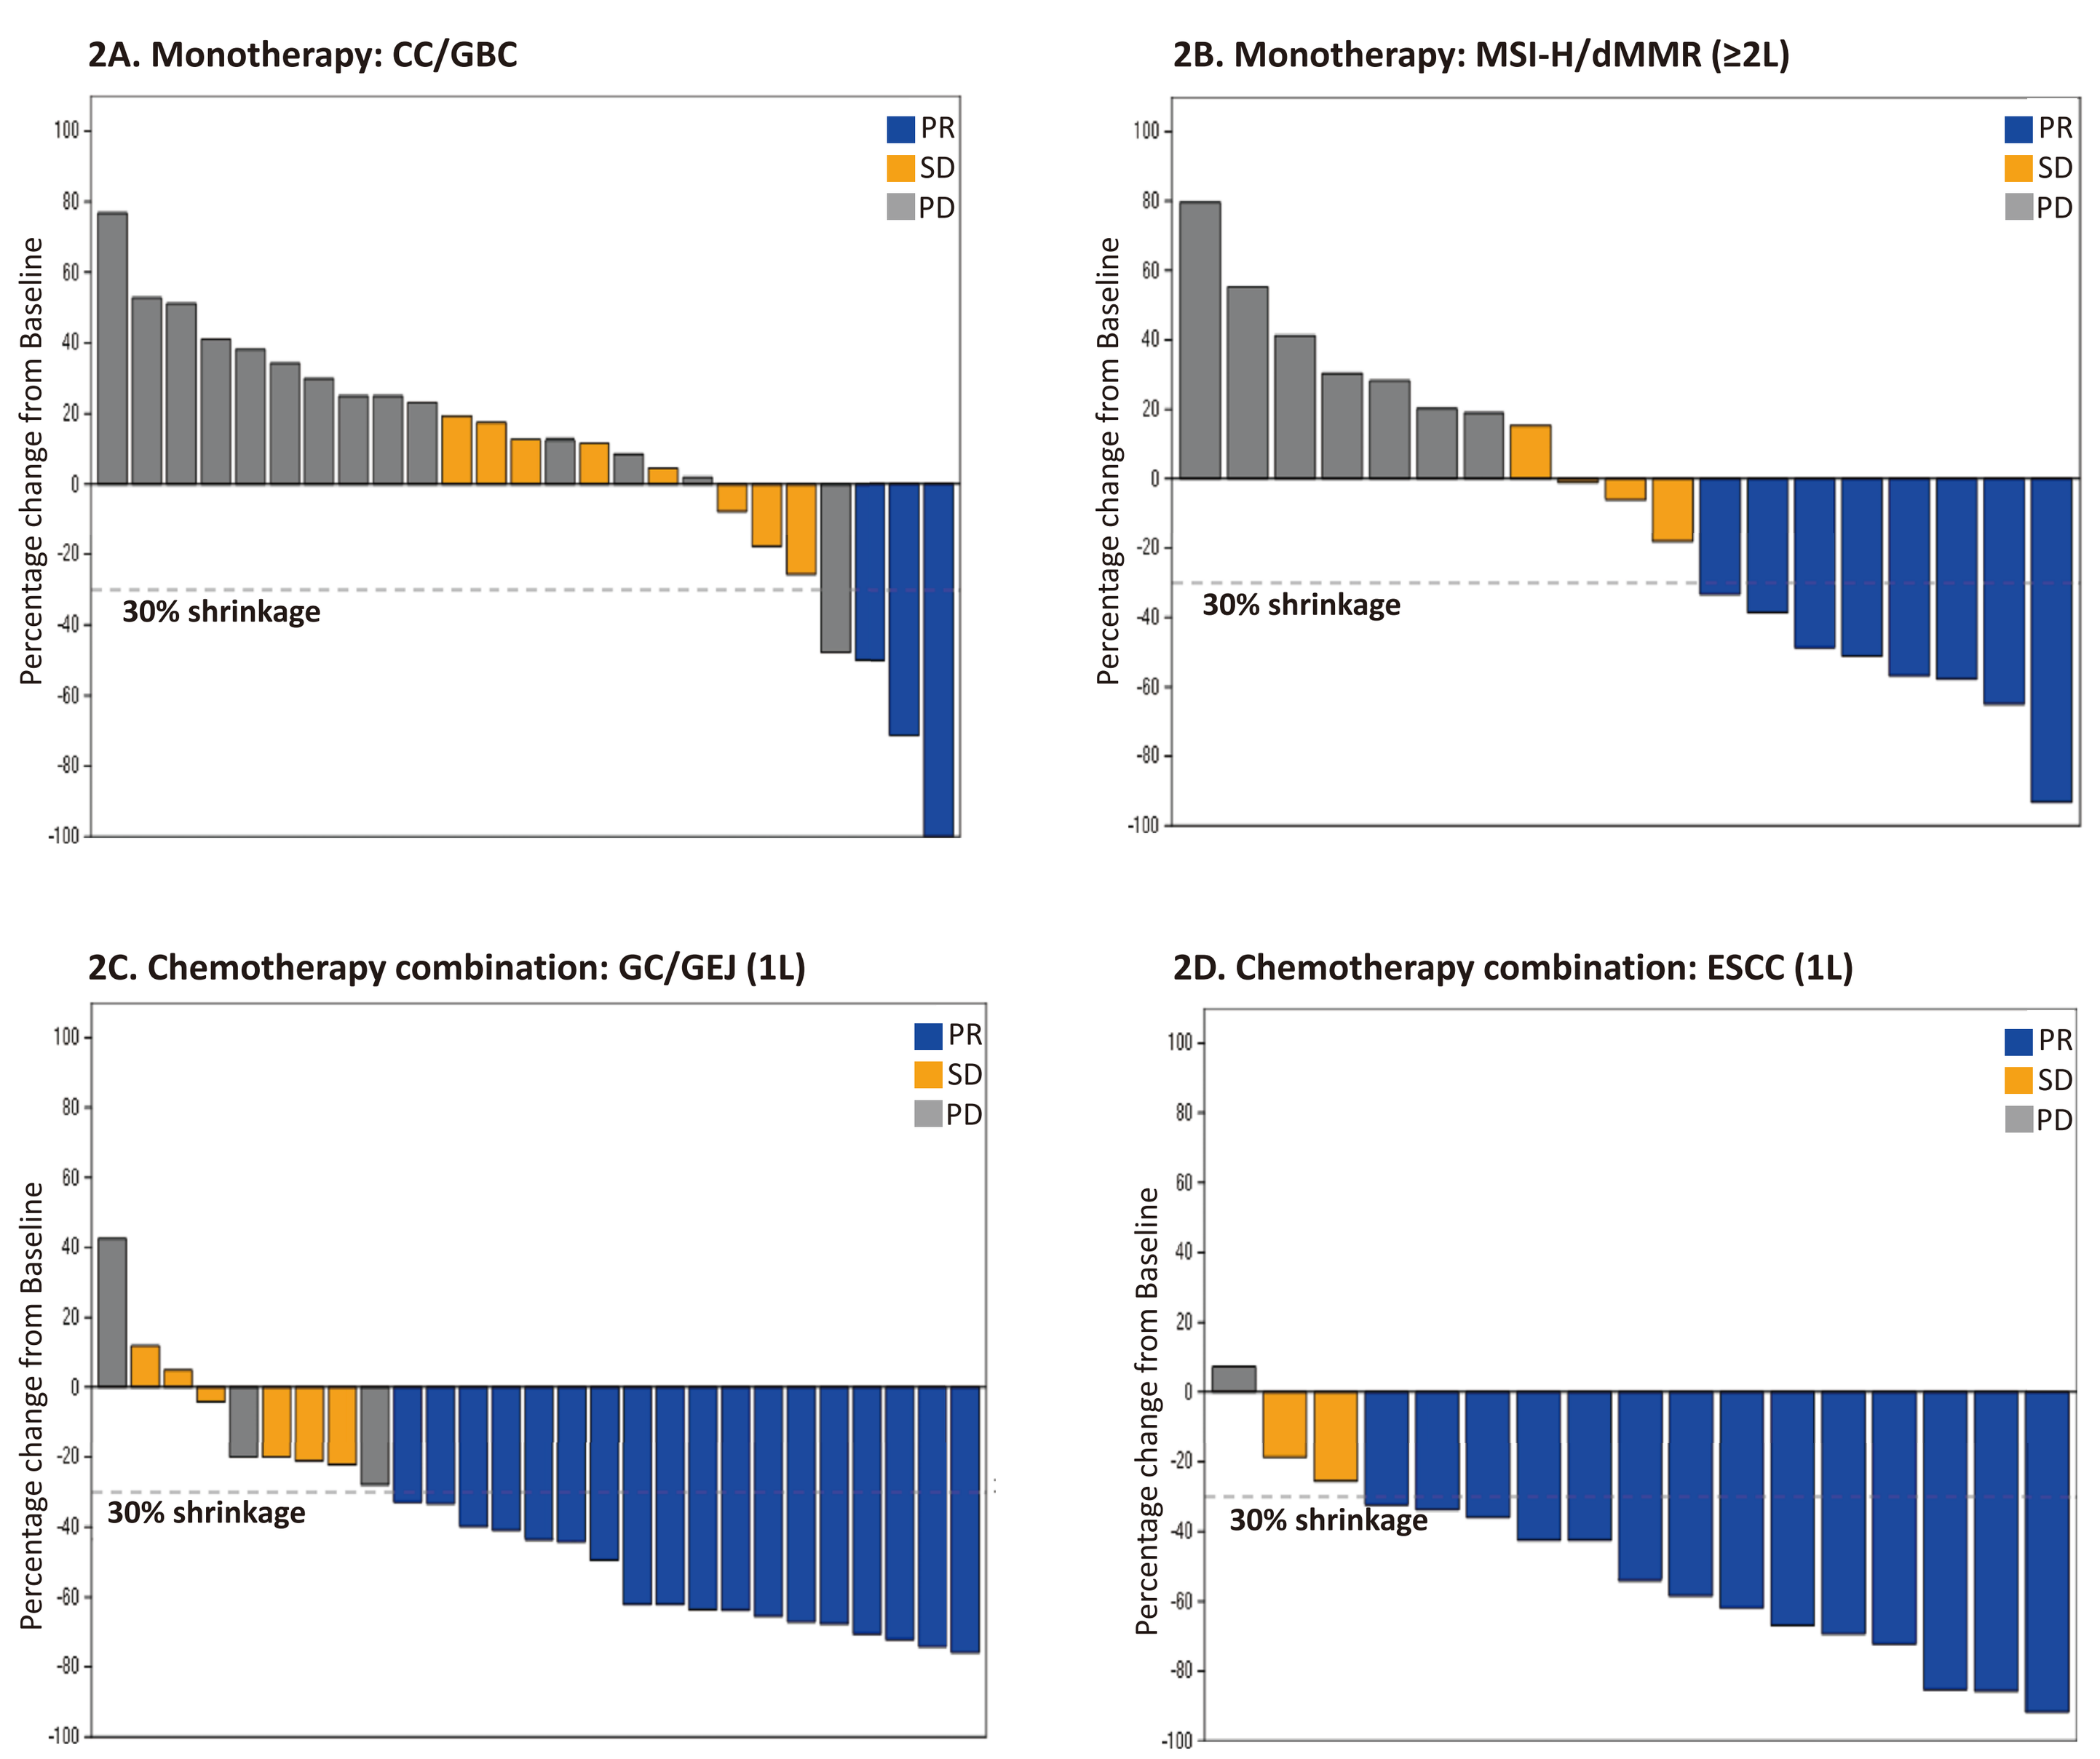
2A. Monotherapy: CC/GBC
2B. Monotherapy: MSI-H/dMMR (≥2L)
2C. Chemotherapy combination: GC/GEJ (1L)
2D. Chemotherapy combination: ESCC (1L)
Percentage change from Baseline
30% shrinkage
PR
SD
PD

Table 2. Summary of Objective Response (Efficacy Analysis Set)- Monotherapy

Response	CC/GBC (N=29) n (%)	MSI-H/dMMR (≥ 2L) (N=21) n (%)
Partial response (PR)	3 (10.3)	8 (38.1)*
Stable disease (SD)	8 (27.6)	4 (19.0)
Progression (PD)	14 (48.3)	7 (33.3)
NA	4 (13.8)	2 (9.5)
ORR	3 (10.3)	8 (38.1)
Confirmed ORR	2 (6.9)**	6 (28.6)
Disease control rate (DCR)	11 (37.9)	12 (57.1)
Duration of response (DoR, Months), Median (Range)	5.39 (1.91+, 8.02)	NR (0.03+, 8.61+)

NA: not applicable; NR: not reached
Four (4) patients and 2 patients in CC/GBC and MSI-H/dMMR cohorts, respectively, did not have any tumor assessment post baseline and were regarded as non-responders (NA in this table), therefore, were not shown in two figures (Figures **2A and 2B**).
*Of the 8 responders in MSI-H/dMMR cohort, 7 patients were colorectal cancer, 1 patient had intestinal cancer
**Of the 3 responders in CC/GBC cohort, 1 achieved first PR after PD and was not counted in confirmed ORR.

Table 3. Summary of Objective Response (Efficacy Analysis Set)- Chemotherapy Combination

Response	GC/GEJ (1L) (N=29) n (%)	ESCC (1L) (N=18) n (%)
Partial response (PR)	18 (62.1)	14 (77.8)
Stable disease (SD)	6 (20.7)	2 (11.1)
Progression (PD)	3 (10.3)	1 (5.6)
NA	2 (6.9)	1 (5.6)
ORR	18 (62.1)	14 (77.8)
Confirmed ORR	15 (51.7)	12 (66.7)*
Disease control rate (DCR)	24 (82.8)	16 (88.9)
Duration of response (DoR, Months) , Median (Range)	6.21 (0.03+, 6.21+)	NR (0.03+, 8.41+)

NA: not applicable; NR: not reached
Two (2) patients and 1 patient in GC/GEJ and ESCC cohorts, respectively, did not have any tumor assessment post baseline and were regarded as non-responders (NA in this table), therefore, were not shown in two figures (Figures **2C and 2D**) .
*Of the 14 responders in ESCC cohort, 2 did not reach the second tumor assessment point by data cut off day and was not counted in confirmed ORR.

Figure 3 Percentage Change from Baseline in Sum of Diameters (Efficacy Analysis Set)

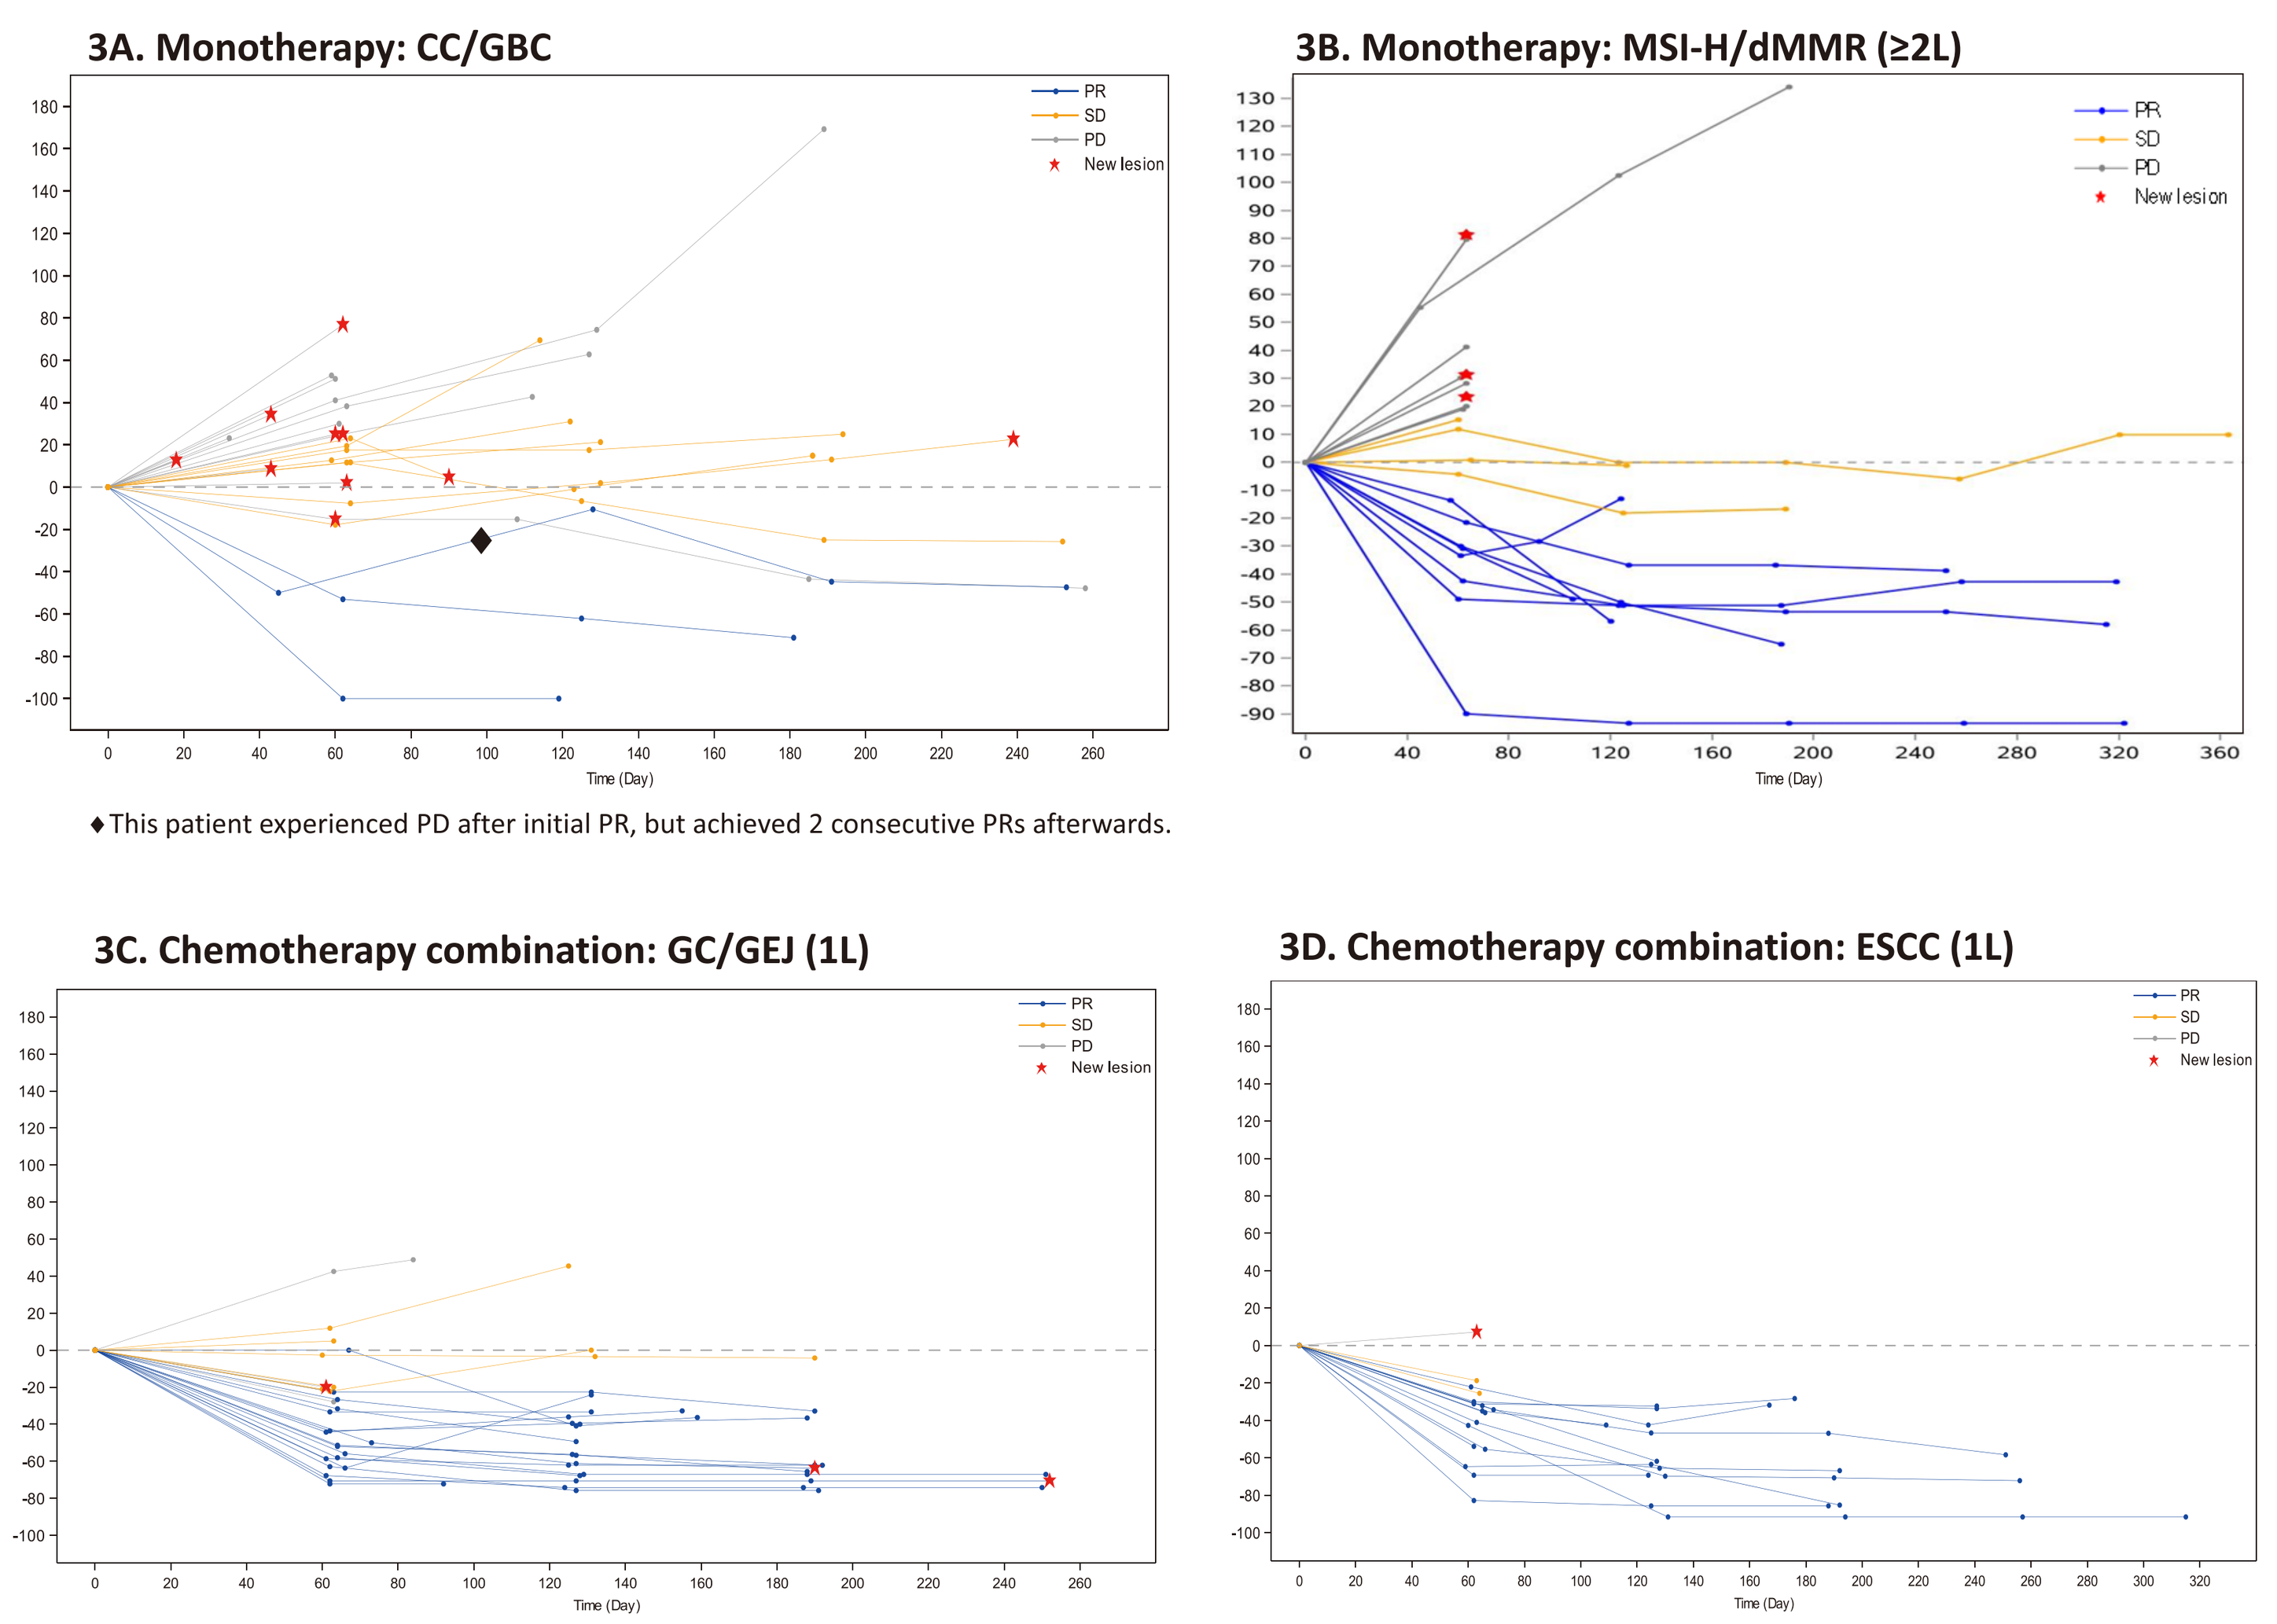
3A. Monotherapy: CC/GBC
3B. Monotherapy: MSI-H/dMMR (≥2L)
3C. Chemotherapy combination: GC/GEJ (1L)
3D. Chemotherapy combination: ESCC (1L)
PR
SD
PD
New lesion
Time (Day)

♦This patient experienced PD after initial PR, but achieved 2 consecutive PRs afterwards.

Figure 4 Duration of Treatment and Tumor Assessment by RECIST V1.1 (Efficacy Analysis Set)

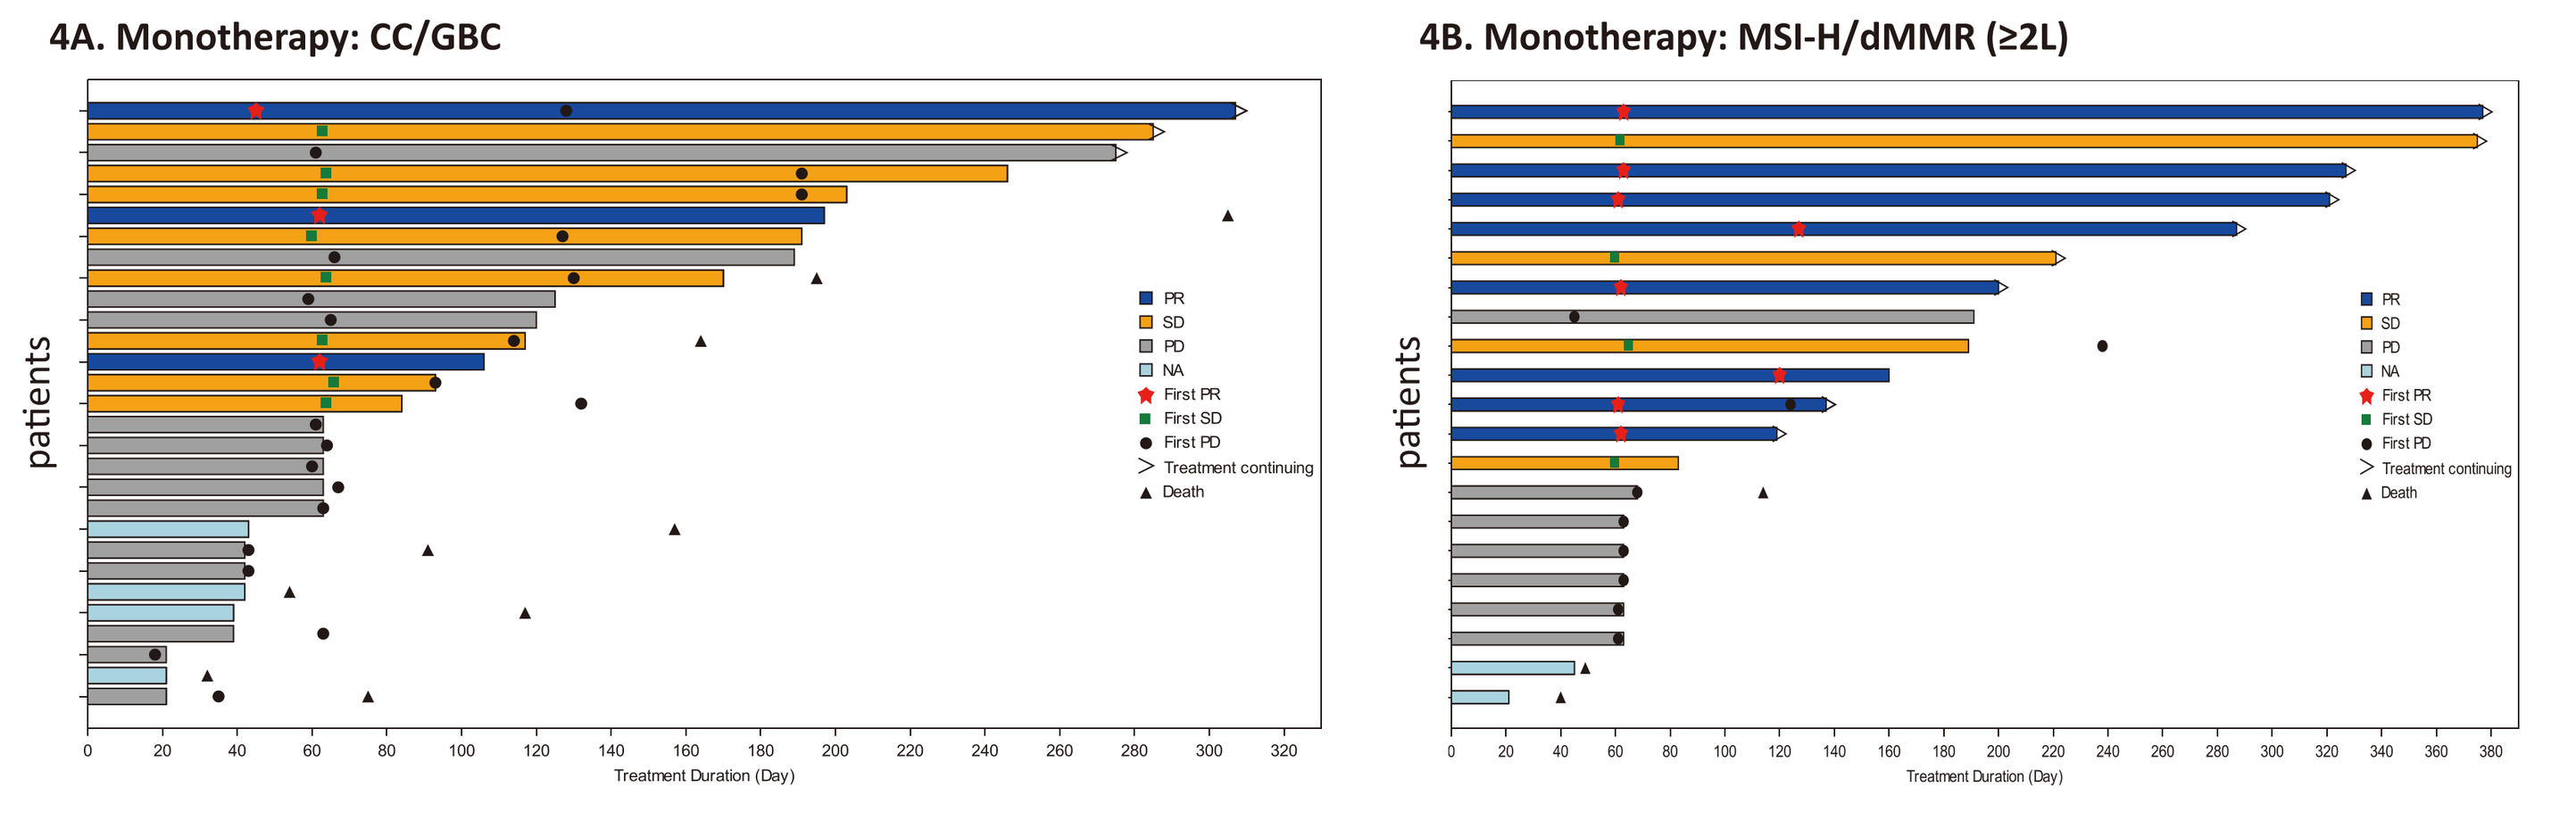
4A. Monotherapy: CC/GBC
4B. Monotherapy: MSI-H/dMMR (≥2L)
patients
Treatment Duration (Day)
PR
SD
PD
NA
First PR
First SD
First PD
Treatment continuing
Death

4A: At the time of data cutoff, 3 patients were still undergoing study treatment, 2 patients had confirmed PR. The median treatment (CS1001) duration was 84 days.

4B: At the time of data cutoff, 9 patients were still undergoing study treatment, 6 patients had confirmed PR. The median treatment (CS1001) duration was 137 days.

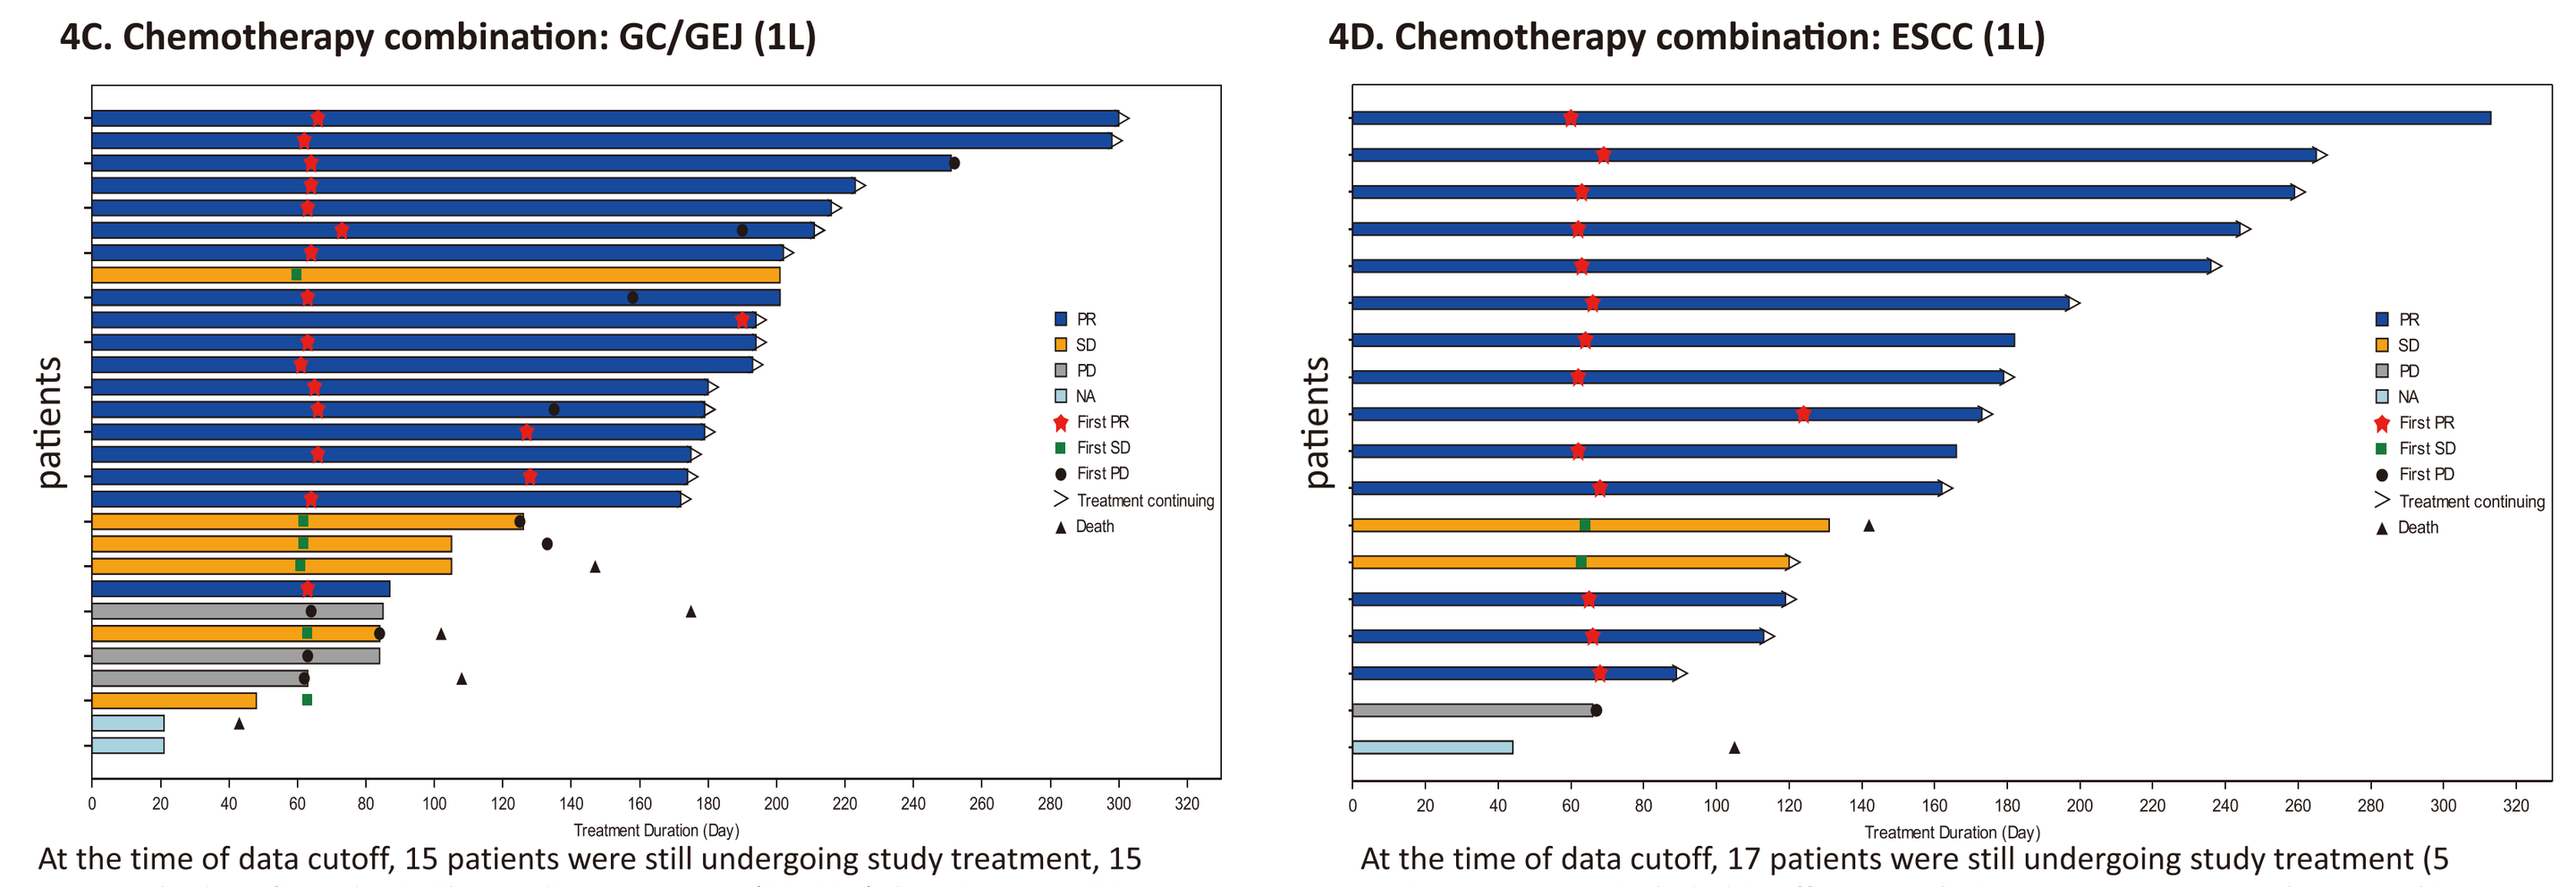
4C. Chemotherapy combination: GC/GEJ (1L)
4D. Chemotherapy combination: ESCC (1L)
patients
Treatment Duration (Day)
PR
SD
PD
NA
First PR
First SD
First PD
Treatment continuing
Death

4C: At the time of data cutoff, 15 patients were still undergoing study treatment, 15 patients had confirmed PR. The median treatment (CS1001) duration was 179 days.

4D: At the time of data cutoff, 17 patients were still undergoing study treatment (5 patients were not included in efficacy analysis set, so as were not shown in this figure), 12 patients had confirmed PR. The median treatment (CS1001) duration was 131 days.

Exposure and Safety - Phase Ib

- As of 01 Jul 2019, 192 patients were enrolled in Phase Ib across multiple cohorts of various tumor types
- Median duration of CS1001 treatment was 112 (Range: 3 - 377) days
- The majority of patients reported treatment-emergent AEs (TEAEs) (monotherapy: 95.9% vs chemotherapy combination: 99.0%). 38.4% of the patients from monotherapy cohorts reported Grade (G) 3-5 TEAEs, and 70.1% of the patients from chemotherapy combination cohorts reported G3-5 TEAEs
- The incidence of serious AEs (SAEs) nearly doubled in the chemo combo cohorts compared to the monotherapy cohorts (40.2% vs 20.5%)

Table 4. Summary of Adverse Events (Safety Analysis Set)

Events	Monotherapy (N=73) n(%)	Chemotherapy Combination (N=97) n(%)
TEAE	70 (95.9)	96 (99.0)
TEAE Related to CS1001	59 (80.8)	87 (89.7)
Grade 3-5 TEAE	28 (38.4)	68 (70.1)
Grade 3-5 TEAE Related to CS1001	11 (15.1)	36 (37.1)
Serious TEAE	15 (20.5)	39 (40.2)
Serious TEAE Related to CS1001	7 (9.6)	22 (22.7)
TEAE Leading to CS1001 Discontinuation	5 (6.8)	8 (8.2)
TEAE Leading to any Combination Chemotherapeutics Withdrawn	NA	20 (20.6)
Immune-related TEAE	34 (46.6)	43 (44.3)
TEAE Leading to Death	0	3 (3.1)
Infusion-related reaction TEAE	1 (1.4)	4 (4.1)

NA: not applicable; TEAE: treatment emergent adverse event

CONCLUSION

- CS1001 monotherapy or combination with standard-of-care chemotherapies demonstrated promising anti-tumor activity and a tolerable safety profile
- Data support further exploration to ongoing development of CS1001 in combination with chemotherapy in GC and ESCC:
 - Phase III trial (GEMSTONE-303, NCT03802591) investigating CS1001 in combination with XELOX in patients with GC/GEJ
 - Phase III trial investigating CS1001 in combination with CF in patients with ESCC
- Data support further exploration and development of CS1001 in CC/GBC and MSI-H/dMMR

REFERENCE

1. Shen, L., et al. Presented at: European Society for Medical Oncology (ESMO), October 19-23, 2018; Munich, Germany, 1165P

ACKNOWLEDGEMENTS

We thank the patients who participated in the study, their families, participating study investigators and clinical sites. This study is sponsored by CStone Pharmaceuticals (Su Zhou) Co., Ltd. Medical writing assistance was provided by Mengxin Chen.

DISCLOSURES

All authors have declared no conflicts of interest.

Poster presented at: *European Society for Medical Oncology (EMSO) – Barcelona, Spain; 27 Sep - 01 Oct 2019 .*
Contact corresponding author: ClinicalDevelopment@cstonepharma.com
ClinicalTrials.gov identifier: NCT03312842

Scan to download a reprint of this poster

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

CStone Pharmaceuticals (Su Zhou) Co., Ltd.