Preliminary Safety, Pharmacokinetics, and Efficacy Results from a Phase I Study of CS1001, an Anti-Programmed Death Ligand-1 Monoclonal Antibody in Patients with Advanced Malignancies (GEMSTONE 101)

MUNICH 2018 ESMO congress
1165P

Shen, Lin[1], Cao, Junning[2], Li, Jin[3], Gong, Jifang[1], Ji, Dongmei[2], Guo, Ye[3], Qin, Zhen[4], Dai, Hangjun[4], Zhang, Zhen[4], Li, Xiao[4], Yang, Jianxin[4]
1. Peking University Cancer Hospital & Institute 2. Fudan University Shanghai Cancer Center 3. Shanghai East Hospital, Tongji University School of Medicine 4. CStone Pharmaceuticals (Su Zhou) Co., Ltd.

基石药业 CSTONE PHARMACEUTICALS

BACKGROUND

- CS1001 is the first full-length, fully human programmed death ligand-1 (PD-L1) targeted immunoglobin G4 (IgG4, s228p) monoclonal antibody (mAb) developed by the OMT transgenic rat platform. The platform mirrors natural IgG4 human antibody, may potentially reduce the risk of immunogenicity and toxicity in patients (pts).
- CS1001 specifically binds to PD-L1, blocking its ligation with programmed cell death protein 1 (PD-1).
- This first-in-human Phase Ia/Ib study was conducted to evaluate the safety, tolerability, pharmacokinetics (PK) profile, and anti-tumor activity of CS1001 in pts with advanced solid tumors or lymphomas.

METHODS

Key Eligibility

Adult pts with histologically or cytologically confirmed metastatic or locally advanced solid tumor or lymphoma; who experienced progression since previous anti-cancer therapy, or for whom standard treatment is not available, not tolerated, or refused (required for Phase Ia); Eastern Cooperative Oncology Group (ECOG) performance status of 0-1.

Study Design

- Pts received CS1001 intravenously once every 3 weeks (Q3W).
- Safety and tolerability were assessed (Phase Ia, 3+3 dose escalation stage). The recommended Phase II dose (RP2D) was determined.
- Pts with various tumor types were enrolled to assess anti-tumor activity and safety in Phase Ib (expansion stage).

Figure 1. Study Design of Phase Ia and Phase Ib

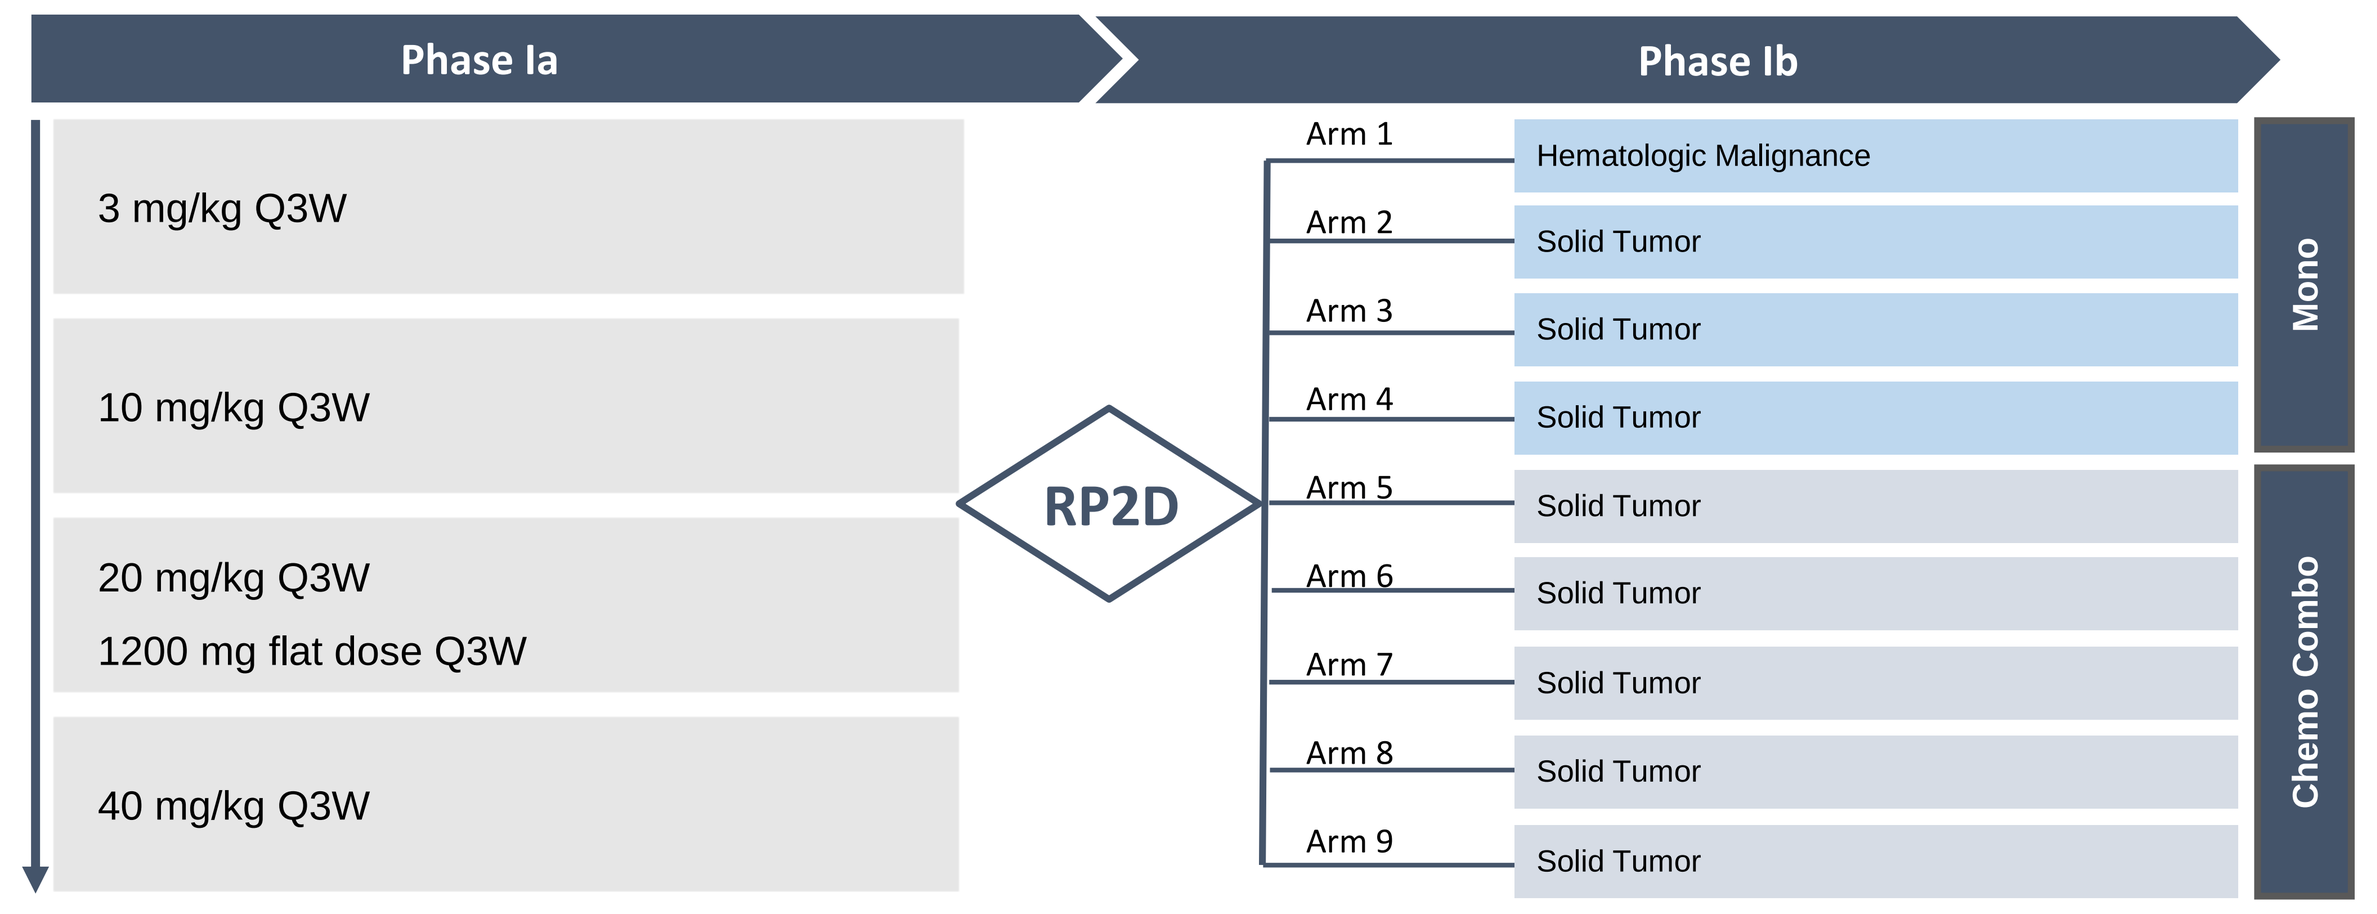

Phase Ia
Phase Ib
3 mg/kg Q3W
10 mg/kg Q3W
20 mg/kg Q3W
1200 mg flat dose Q3W
40 mg/kg Q3W
RP2D
Arm 1 Hematologic Malignance
Arm 2 Solid Tumor
Arm 3 Solid Tumor
Arm 4 Solid Tumor
Arm 5 Solid Tumor
Arm 6 Solid Tumor
Arm 7 Solid Tumor
Arm 8 Solid Tumor
Arm 9 Solid Tumor
Mono
Chemo Combo

Assessments

- Adverse events (AEs) were graded according to the National Cancer Institute Common Terminology Criteria for AE (NCI-CTCAE) v4.03.
- Tolerability evaluations include assessing dose-limiting toxicities (DLTs) during the first cycle.
- Blood specimens were collected for PK analysis at predefined timepoints. PK parameters were derived from non-compartmental analysis from the serum concentration-time profile of CS1001.
- Immunogenicity was characterized by assessing the level of anti-drug antibody (ADA) in the pts.
- Tumor assessments were performed using RECIST v1.1 Q9W (for solid tumors) and Lugano 2014 Q12W (for lymphomas).

RESULTS

Baseline Characteristics

- At the time of data cut-off (July 20, 2018), 29 pts were enrolled in Phase Ia, and 19 pts were enrolled in Phase Ib (Table 1).
- Thirty-five (35) pts are still undergoing study treatment (Phase Ia, n=18; Phase Ib, n=17).
- In Phase Ia, 11 pts have discontinued treatment: 8 due to disease progression, 1 each due to lost to follow-up, death, and other. In Phase Ib, 2 pts have discontinued treatment, both due to disease progression.

Table 1. Patient Demographics and Baseline Characteristics

Characteristic (Unit)	Phase Ia Total (N=29)	Phase Ib Total (N=19)
Age (yr), median (range)	53 (23-75)	49 (32-68)
Sex: Male	19 (65.6)	13 (68.4)
Female	10 (34.5)	6 (31.6)
ECOG performance status:		
0	4 (13.8)	1 (5.3)
1	25 (86.2)	18 (94.7)
Prior anti-cancer therapy line, median (range)	2 (0-7)[a]	2 (0-9)
Treatment duration (days), median (range)	87 (21-275+)	22 (3+-67+)

ECOG: Eastern Cooperative Oncology Group.
a. Two (2) pts had no prior anti-cancer treatment. One (1) had Stage IV abdominal wall sarcoma, no standard therapy available. The other is a 74 year-old rectal adenocarcinoma patient with MSI-H, assessed as not tolerable of chemotherapy.

Safety and Tolerability -- *Phase Ia*

- The median duration of treatment at the cut-off date was 87 days (range: 21 to 275+ days).
- No dose-limiting toxicities (DLTs) were observed. The maximum tolerated dose (MTD) was not reached.
- The most frequent treatment-related AEs (TRAEs), shown in Table 2, were anaemia (41%), proteinuria (24%), blood bilirubin increase (21%). All of them are of CTCAE Grade (G) 1-2, except for 1 G3 anaemia.
- Nine (9) G1-2 immune-related AEs (irAEs) were reported in 4 pts, including endocrine disorders (10%), skin and subcutaneous tissue disorders (7%), and investigations (3%).
- Four (4) serious AEs (SAEs) were reported, which were assessed as not related to the treatment. No treatment-related death were observed.

Table 2. Treatment-Related Adverse Events (TRAEs) Occurred in ≥ 10% Patients or Grade ≥3

Preferred Term	Phase Ia Total (N=29)		Phase Ib Total (N=17)[a]	
	Any Grade	Grade ≥3	Any Grade	Grade ≥3
Number of pts with at least 1 TRAE	24 (82.8)	2 (6.9)	10 (58.8)	0
Anaemia	12 (41.4)	1 (3.5)	2 (11.8)	0
Proteinuria	7 (24.1)	0	2 (11.8)	0
Blood bilirubin increased	6 (20.7)	0	2 (11.8)	0
Aspartate aminotransferase increased	5 (17.2)	0	0	0
Decreased appetite	5 (17.2)	0	0	0
Alanine aminotransferase increased	4 (13.8)	0	0	0
Bilirubin conjugated increased	4 (13.8)	0	2 (11.8)	0
White blood cell count decreased	4 (13.8)	0	0	0
Nausea	4 (13.8)	0	0	0
Rash	3 (10.3)	0	0	0
Platelet count decreased	1 (3.5)	1 (3.5)	0	0
Blood creatine phosphokinase MB increased	0	0	2 (11.8)	0

a. Excluded 2 pts from CS1001 and chemotherapy combination arm (Arm 5). No TRAEs were observed in this arm at the time of data cut-off.

Safety and Tolerability -- *Phase Ib*

- The median duration of treatment at the cut-off date was 87 days (range: 3+ to 67+ days).
- The most frequent TRAEs in pts from monotherapy arms (Arm 1-4), shown in Table 2, were G1-2 anaemia (12%), proteinuria (12%), blood bilirubin increase (12%), bilirubin conjugated increased (12%), and blood creatine phosphokinase MB increased (12%).
- Twelve (12) G1-2 irAEs were reported in 5 pts from monotherapy arms (Arm 1-4).
- Two (2) SAEs were reported, which were assessed as not related to the treatment, no treatment-related death were observed (Arm 1-4).
- No TRAEs, irAEs or SAEs were reported in pts from CS1001 combination chemotherapy arm (Arm 5).

Pharmacokinetics and Immunogenicity

PK data from 29 pts of Phase Ia (Cycle 1) demonstrated that:

- CS1001 had a dose-proportional PK profile across 5 dose levels.
- Clearance (CL) was 0.1-0.3 L/day; $T_{1/2}$ was 12-17 days.

Immunogenicity data from 29 pt of Phase Ia demonstrated that:

- Seven (7) pts had at least 1 post-baseline positive ADA results. The incidence of ADA was 24%.

Figure 2. CS1001 PK profile for All Doses (Cycle 1)

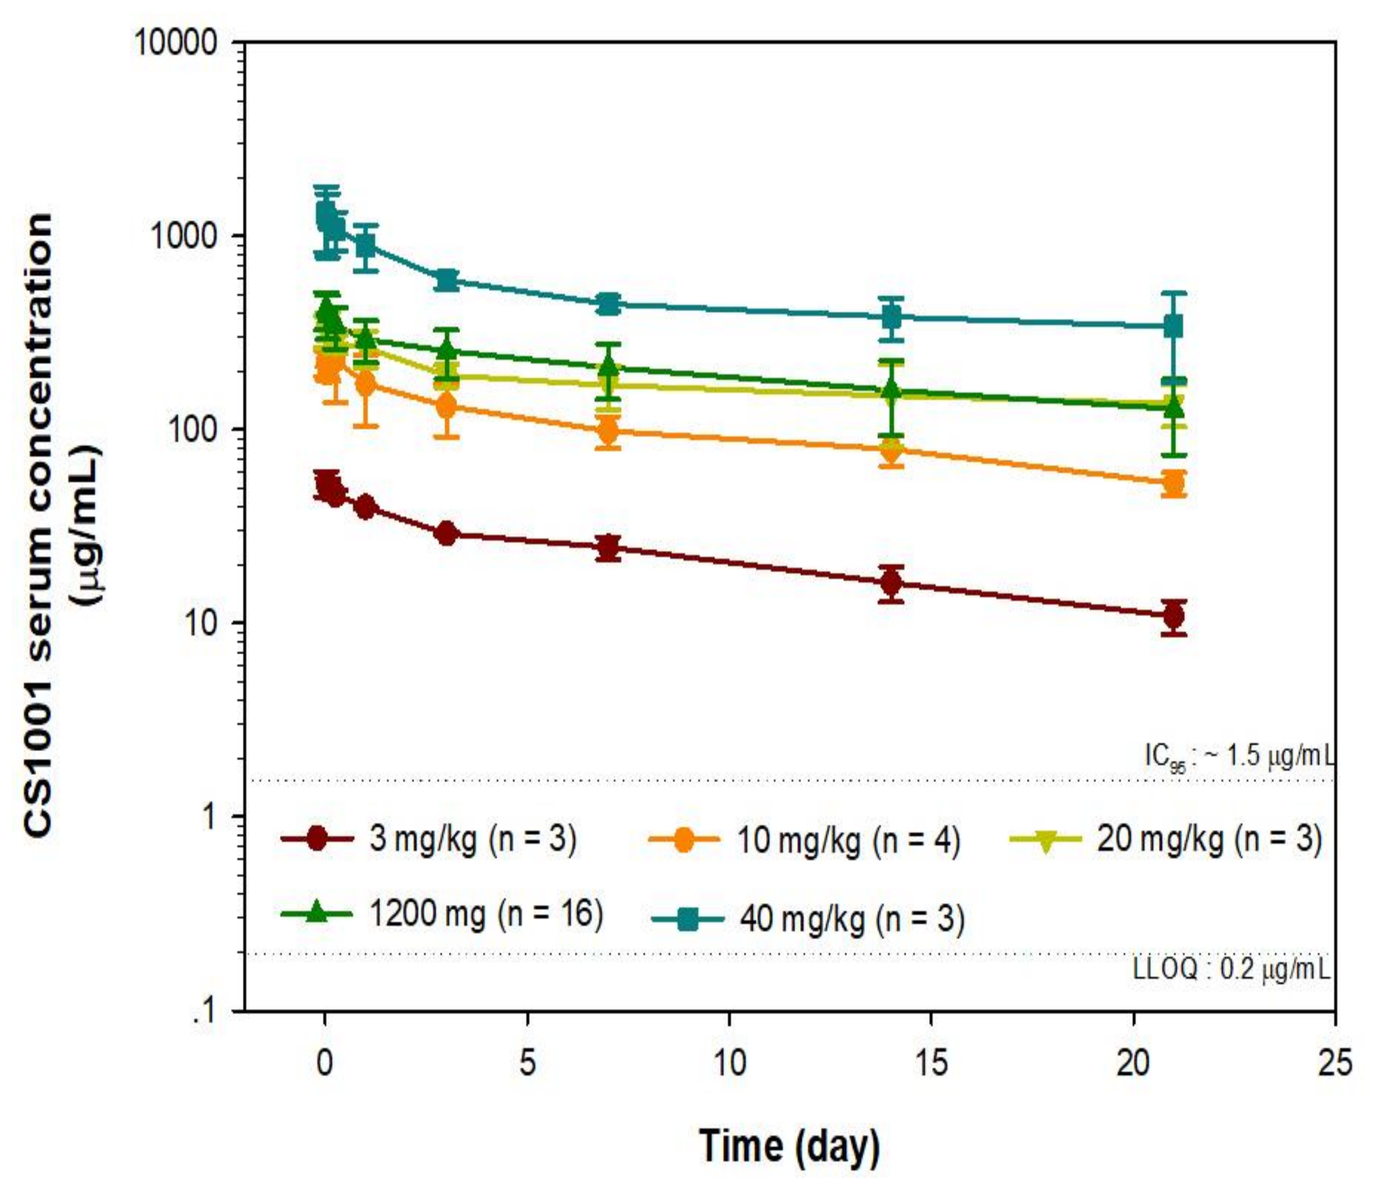

CS1001 serum concentration (μg/mL)
10000
1000
100
10
1
.1
0 5 10 15 20 25
Time (day)
3 mg/kg (n = 3)
10 mg/kg (n = 4)
20 mg/kg (n = 3)
1200 mg (n = 16)
40 mg/kg (n = 3)
LLOQ : 0.2 μg/mL

IC_{95} of *in vitro* binding assay for blocking ligation of PD-L1 with PD-1 was (1.51 μg/mL)

Efficacy

- In Phase Ia, 5 (20%) pts achieved partial response (PR), 8 (32%) pts achieved stable disease (SD). The median treatment duration for these patients was 178 days (range: 64+ to 275+).
- At the time of data cut-off (July 20, 2018), due to limited follow-up of Phase Ib, the efficacy results are not summarized.

Figure 3. Best Objective Responses (BOR) in Phase Ia

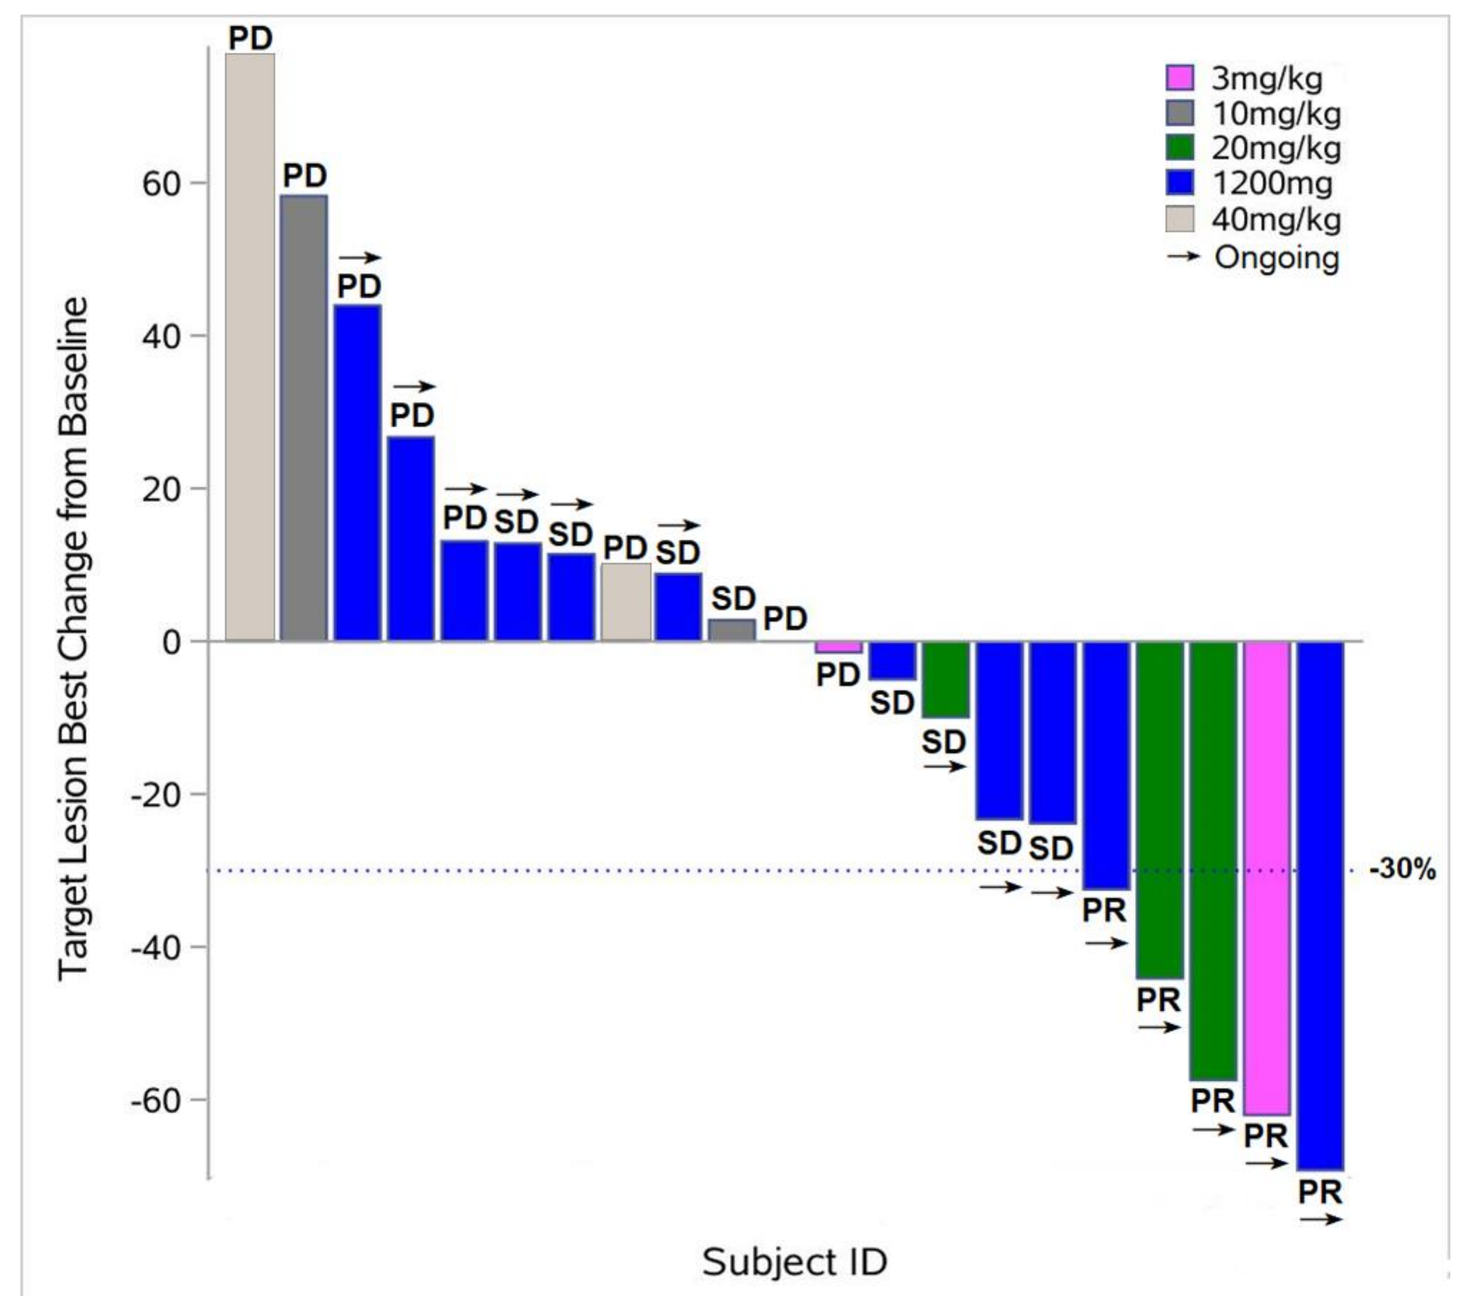

Target Lesion Best Change from Baseline
60
40
20
0
-20
-40
-60
-30%
Subject ID
3mg/kg
10mg/kg
20mg/kg
1200mg
40mg/kg
Ongoing

Response，n (%)	Total (N=25*)
Partial response (PR)	5 (20.0)
Stable disease (SD)	8 (32.0)
Progressive disease (PD)	9 (36.0)
Objective response (CR+PR)	5** (20.0)
Disease control (CR+PR+SD)	13 (52.0)

Patients with PR: ampullary carcinoma with MSI-H, cholangiocarcinoma, NSCLC, cervical cancer, mixed histology of esophagus cancer and melanoma
*Twenty-five (25) pts were included in the efficacy analysis set, which is defined as pts who received study drug and had measurable disease at baseline (4 ongoing pts who had not reached the 1st post-baseline tumor assessment were excluded). Of the 25 pts, 4 were not shown on the plot due to no post-baseline target lesion evaluation: 3 pts had the BOR of not evaluable, and 1 had PD
** Response include 4 confirmed and 1 unconfirmed response, but subsequently confirmed after the cut-off date

Figure 4. Duration of Treatment in Phase Ia

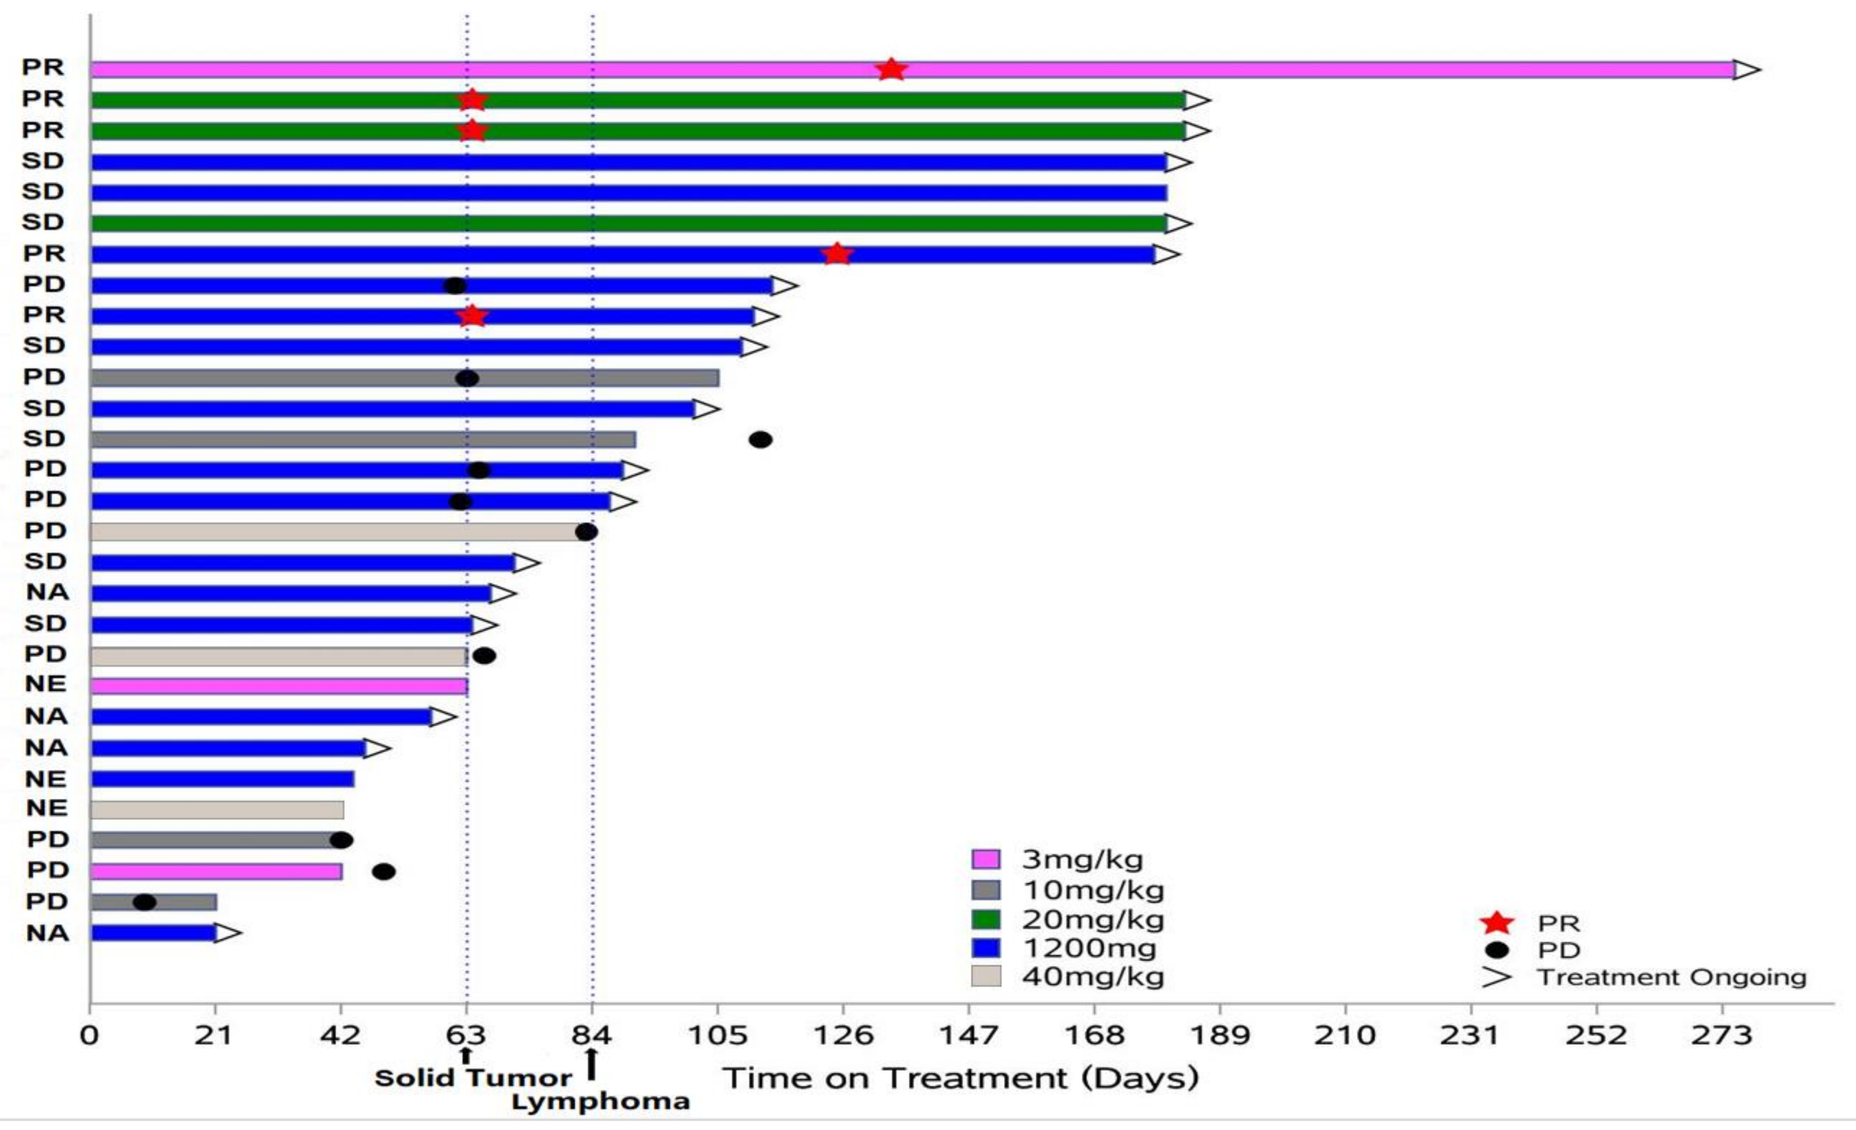

3mg/kg
10mg/kg
20mg/kg
1200mg
40mg/kg
PR
PD
Treatment Ongoing
0 21 42 63 84 105 126 147 168 189 210 231 252 273
Solid Tumor
Lymphoma
Time on Treatment (Days)

- As of July 20, 2018, the majority of patients (62.1%; n=18/29) are still on treatment
- Among patients with PR and SD, median treatment duration was 178 days (range: 64+ to 275+ days)

*Tumor assessment performed every 9 weeks for solid tumors and every 12 weeks for lymphomas
**PR: partial response; PD: progressive disease; SD: stable disease; NE: not evaluable; NA: not assessed

Figure 5. Response to CS1001 in Patient, Stage IV MSI-H Ampullary Carcinoma with Retroperitoneal Lymph Node Metastases

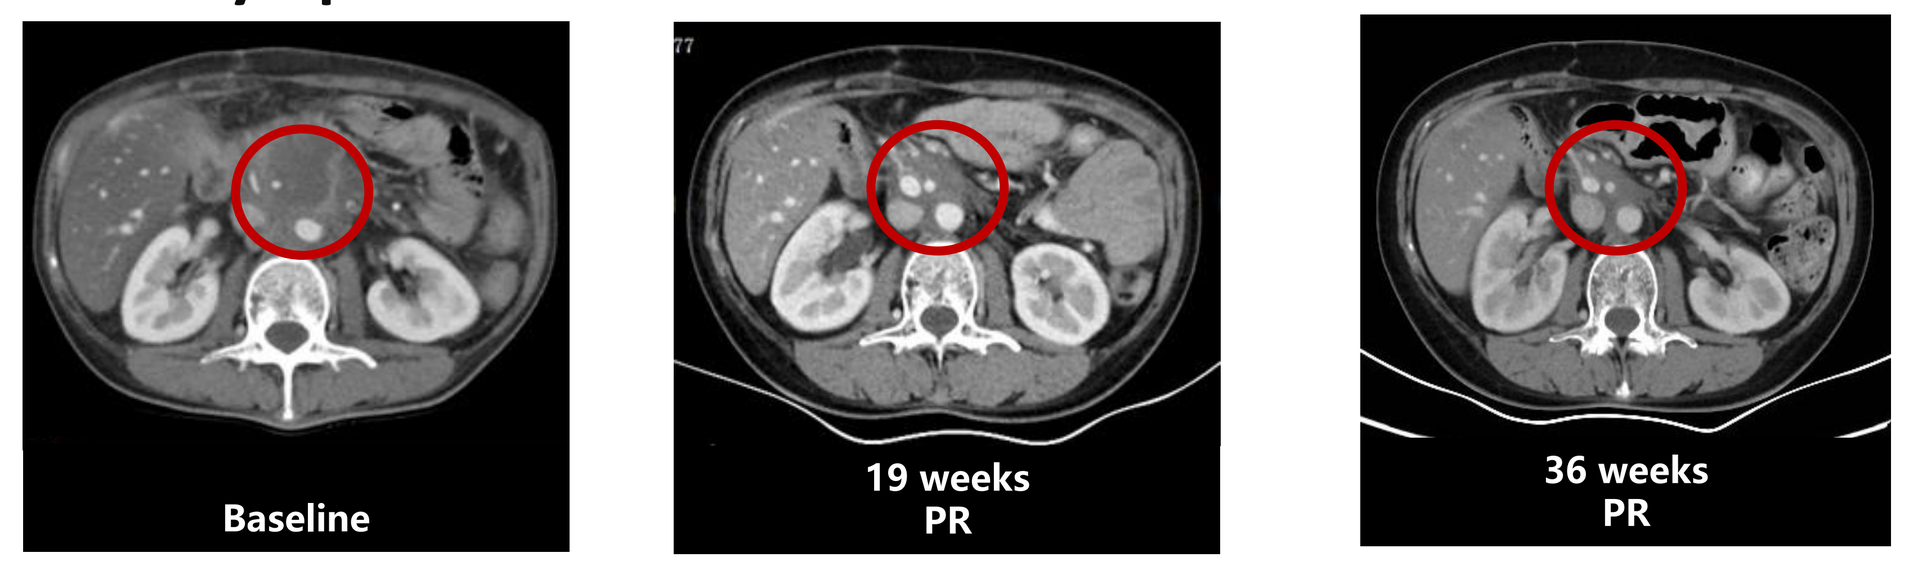

Baseline
19 weeks PR
36 weeks PR

3 mg/kg Q3W cohort; ongoing response at the time of data cut-off

Figure 6. Response to CS1001 in Patient, Stage IV NSCLC and Adenocarcinoma with Multiple Extrathoracic Metastases

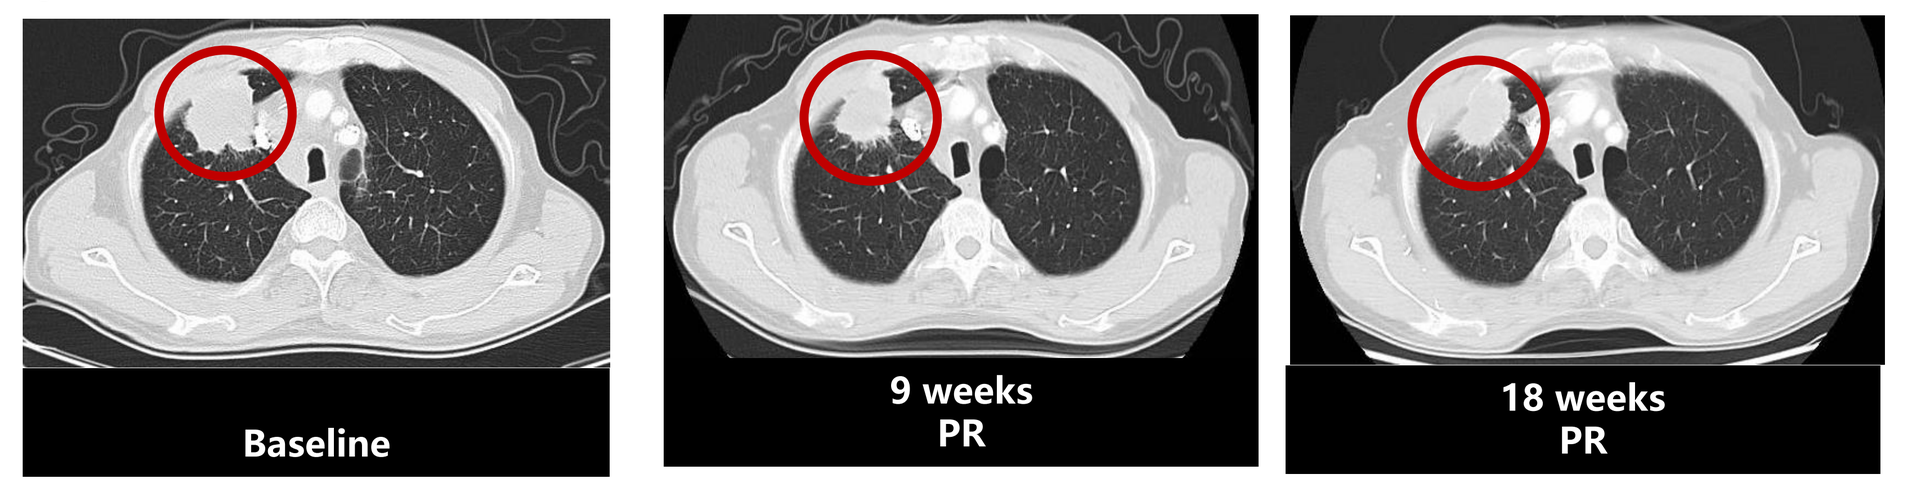

Baseline
9 weeks PR
18 weeks PR

20 mg/kg Q3W cohort; ongoing response at the time of data cut-off

CONCLUSION

- CS1001 was well tolerated with no reported DLT and drug-related SAE.
- Early anti-tumor activity observed among 25 patients, including 5 PRs (all remain on treatment).
- CS1001 demonstrated dose-proportional PK profile with $T_{1/2}$ of 12~17 days.
- 1200 mg Q3W flat dose determined as RP2D based on safety, PK, and preclinical pharmacology data.
- The preliminary safety profile and antitumor activity support continued development of CS1001 in patients with advanced tumors.

ACKNOWLEDGEMENTS

We thank the patients who participated in the study, their families, participating study investigators and clinical sites. This study is sponsored by CStone Pharmaceuticals (Su Zhou) Co., Ltd

Poster presented at: *European Society for Medical Oncology (EMSO) – Munich Germany; October 19-23, 2018.*

Contact corresponding author: ClinicalDevelopment@cstonepharma.com

ClinicalTrials.gov identifier: NCT03312842

Scan to download a reprint of this poster
Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

CStone Pharmaceuticals (Su Zhou) Co., Ltd.